#2181

INTRAMUSCULAR IBALIZUMAB: PHARMACOKINETICS, SAFETY, AND EFFICACY VS IV ADMINISTRATION

Hsi-Hsun Lin[1], Susan Shin-Jung Lee[2], Ning-Chi Wang[3], YingAn Lai[4], Kuei-Ling Kuo[4], Steven Weinheimer[5], Stanley Lewis[5]

[11]E-Da Hospital/I-Shou University, Kaohsiung City, Taiwan, [2]Kaohsiung Veterans General Hospital, Kaohsiung, Taiwan,
[3]Tri-Service General Hospital, National Defense Medical Center, Taipei City, Taiwan, [4]TaiMed Biologics, Taipei City, Taiwan, [5]TaiMed Biologics USA, Irvine, CA, USA

Background

Ibalizumab (IBA) is a long-acting humanized monoclonal antibody that binds to the second extracellular domain of the CD4 receptor. It is currently in Phase 3 development for treatment of multi-drug resistant (MDR) HIV-1 infection. IBA has been safely administered intravenously (IV) and subcutaneously (SC) in previous clinical trials. In this ongoing adaptive design study, we aimed to investigate safety, pharmacokinetics (PK), antiviral activity, and pharmacodynamics (PD) of intramuscular (IM) administration. This report is limited to results of the 800 mg and 2000 mg IM dose groups as it offers direct comparison to the previous Phase 2 IV administration trial in patients with MDR HIV which studied the same dosages.

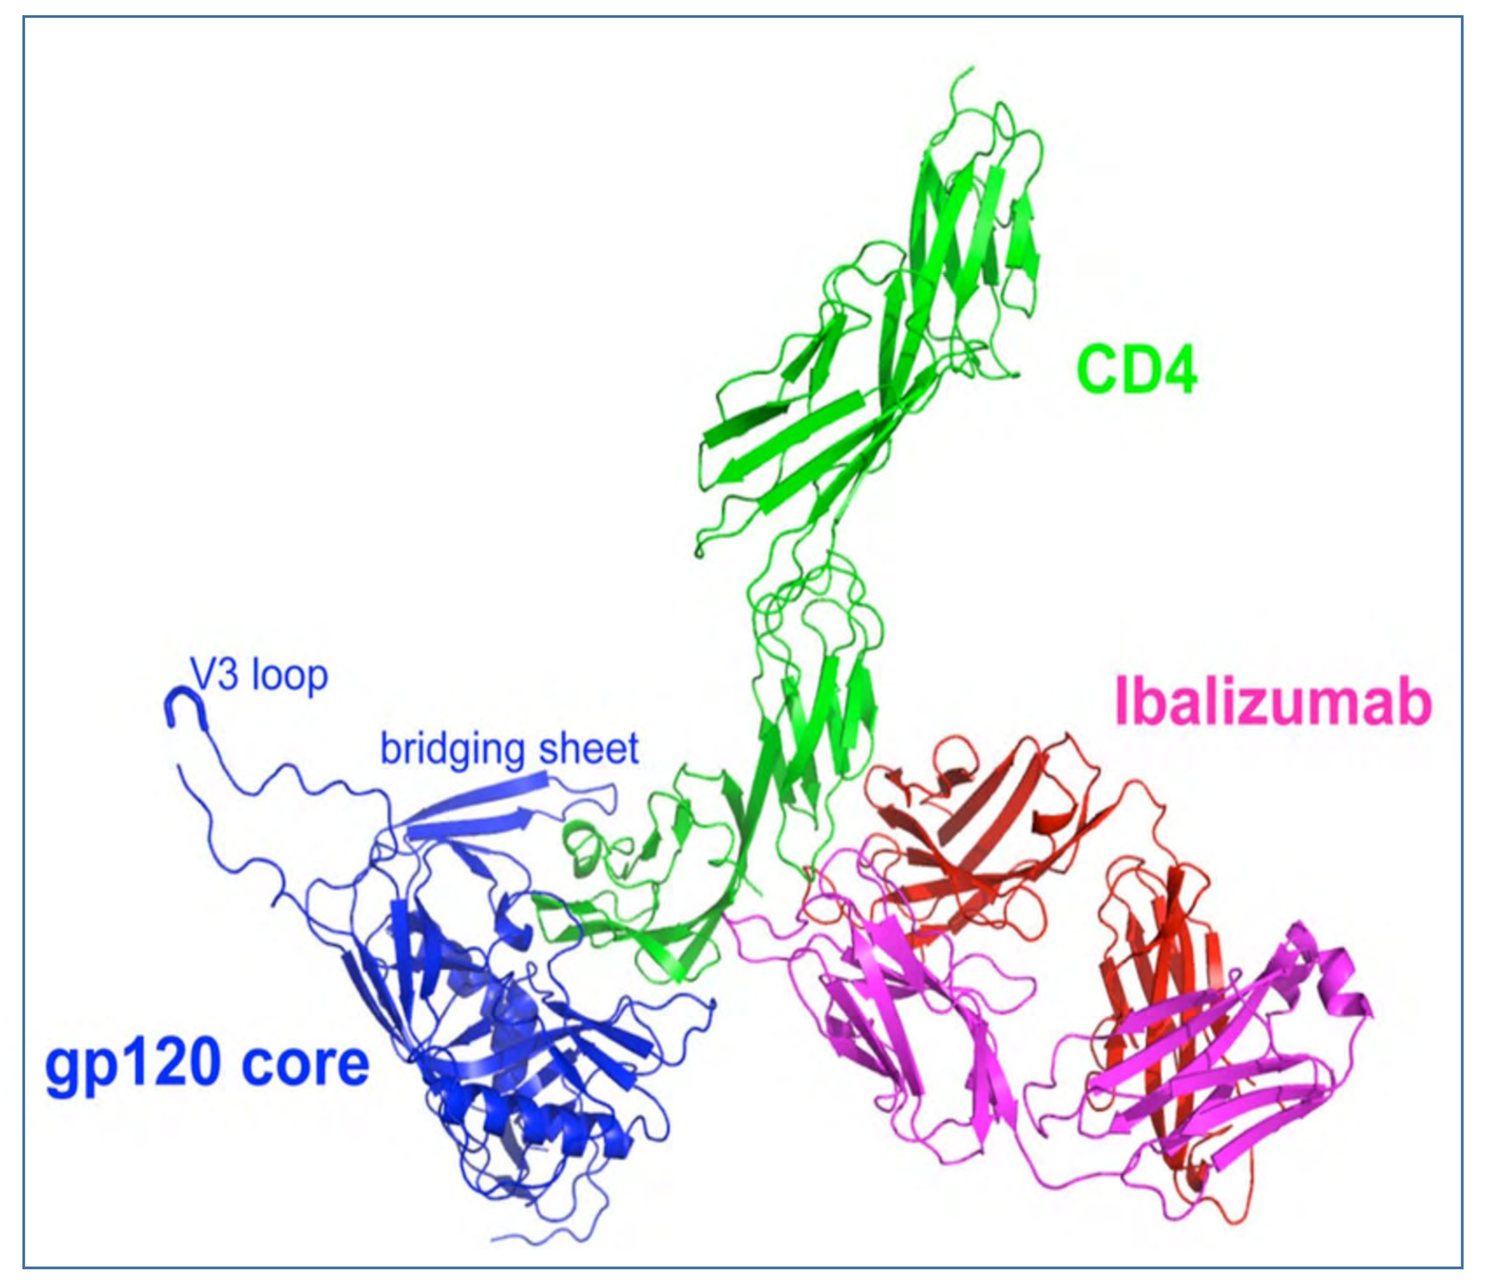
CD4
V3 loop
bridging sheet
Ibalizumab
gp120 core

Methods

TMB-121: Phase 1/2, randomized, placebo-controlled study of IBA SC/IM injection

- Cohort 1: HIV-infected patients with HIV-1 RNA ≥5000 copies and who have not received antiretroviral (ARV) treatment for at least one year
- Cohort 1 Arms E and F: 100% male, 100% Asian, mean age 27 years, mean weight 64kg, mean baseline viral load (VL) 57,000 copies/mL, mean baseline CD4$^+$ T cell count 336

Cohort/ Arm	Route	Treatment Regimen	Study Duration (Week)	Number of Subjects
1/E	IM	800 mg (5.33 mL) at Days 0, 14 and 28	8	8
1/F	IM	2000 mg (13.33 mL) at Days 0 and 28	10	6

TMB-202: Phase 2b, randomized, double-blinded study of IBA IV injection

- HIV-infected patients with HIV-1 RNA >1000 copies and who have been receiving HAART for at least 6 months and were to be failing, or were to have recently failed (i.e., in the last 8 weeks) therapy
- 89% male, 62% White and 24% Black, mean age 48 years, mean weight 81 kg, mean baseline VL 125,000 copies/mL, mean baseline CD4$^+$ T cell count 109
- The two dose regimens were randomly assigned in a 1:1 ratio.

Arm	Route	Treatment Regimen	Study Duration (Week)	Number of Subjects
A	IV	800 mg every 2 weeks plus OBR for 24 weeks	28	59
B	IV	2000 mg every 4 weeks plus OBR for 24 weeks	28	54

Pharmacokinetics

800 mg IBA - IM vs. IV)

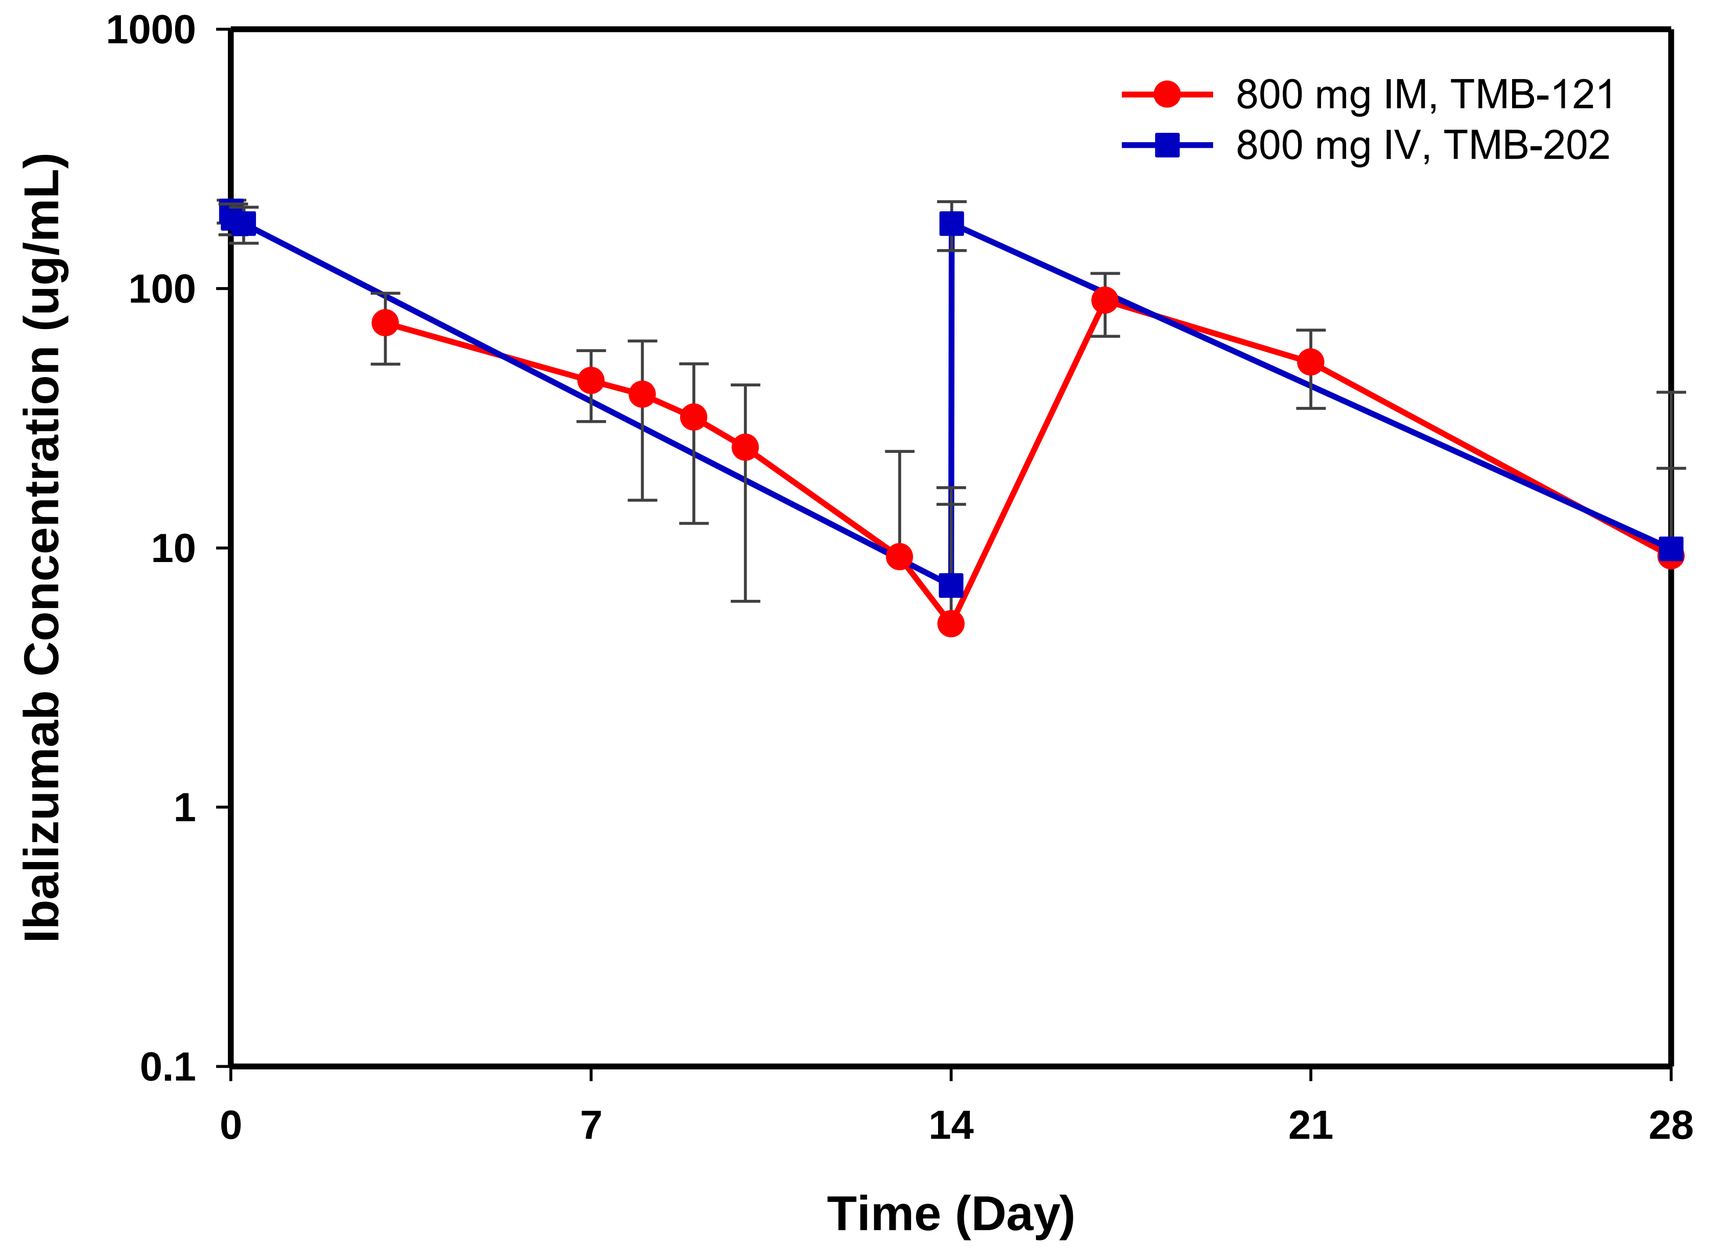
800 mg IM, TMB-121
800 mg IV, TMB-202
Ibalizumab Concentration (ug/mL)
Time (Day)

2000 mg IBA - IM vs. IV)

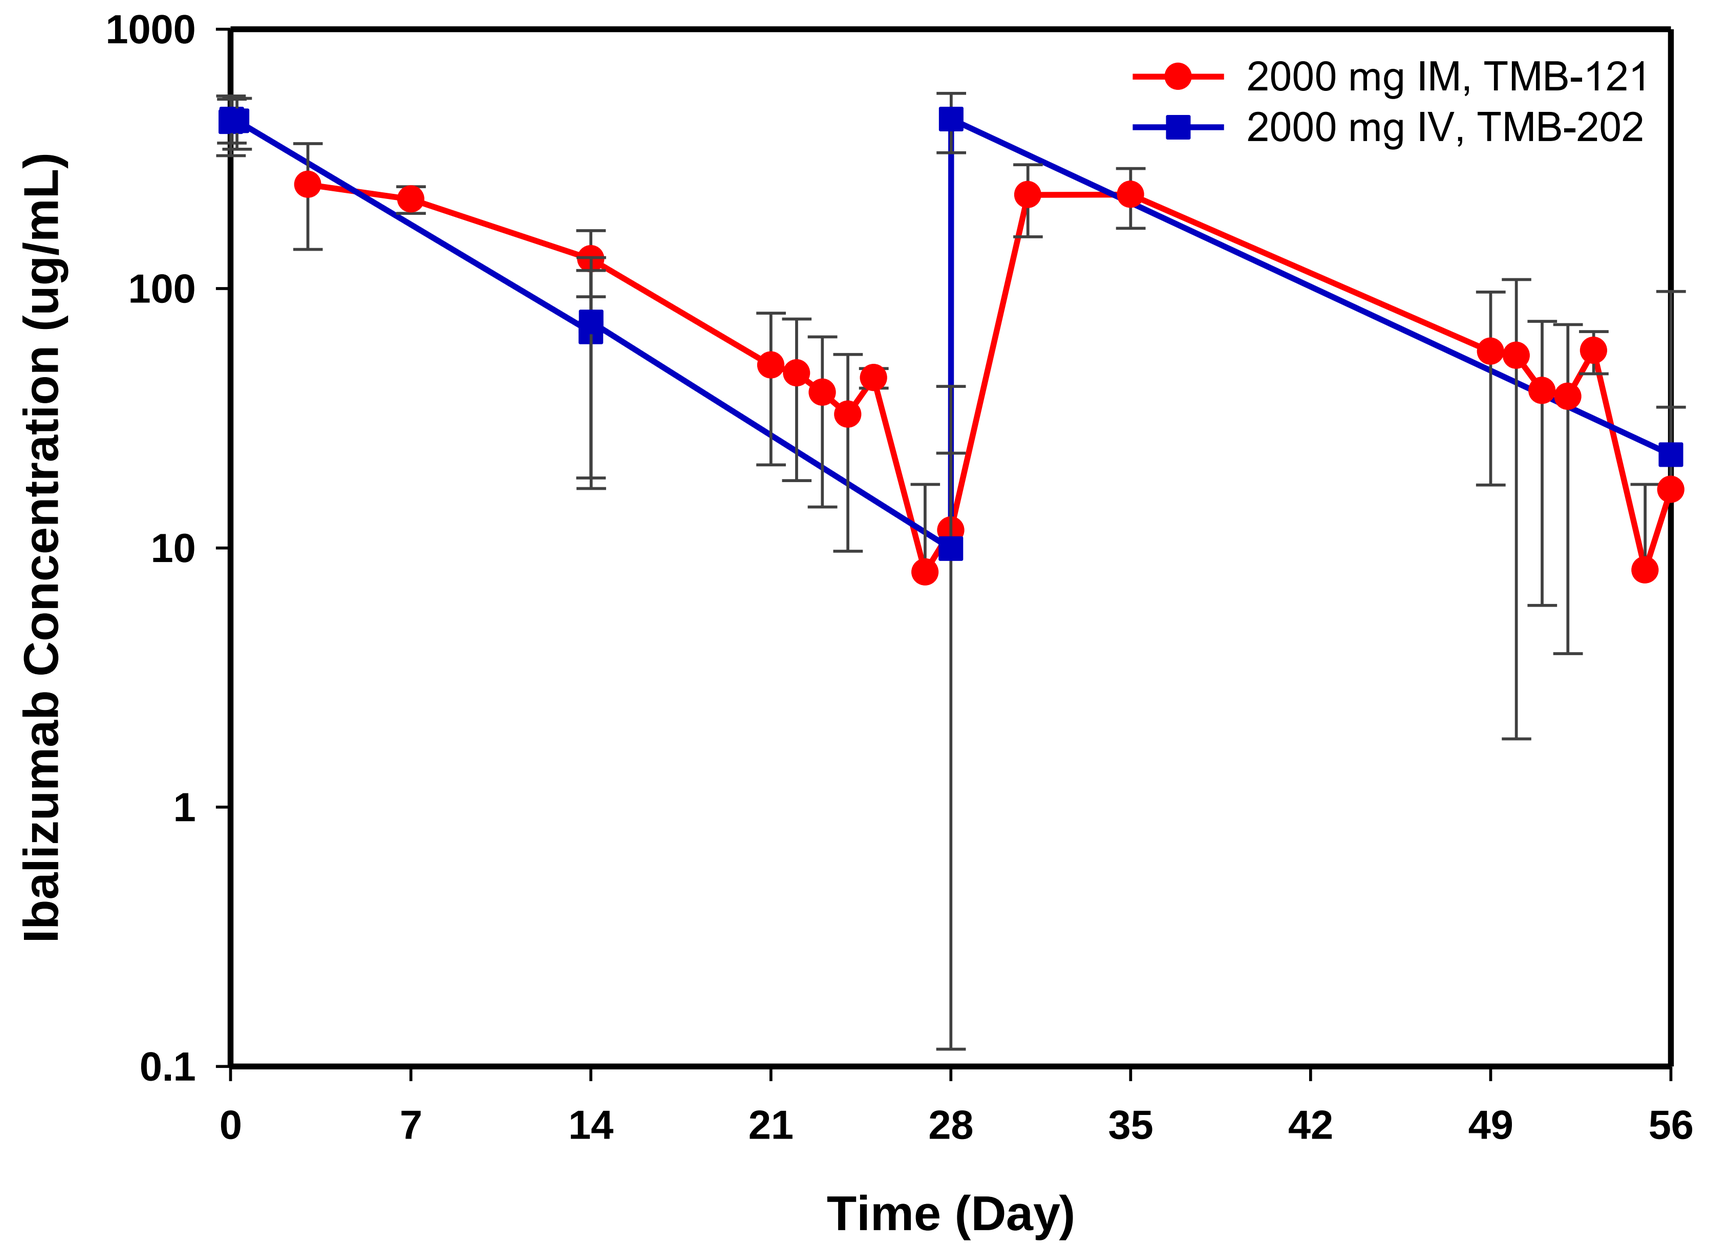
2000 mg IM, TMB-121
2000 mg IV, TMB-202
Ibalizumab Concentration (ug/mL)
Time (Day)

- The PK profiles of 800 mg and 2000 mg IM IBA injection were comparable with IV IBA injections while maintaining effective concentrations during the dosing period.

PK Parameters, Mean (SD)

Dose (mg)	Route	No. of dose	T_{max} (day)	C_{max} (µg/mL)	AUC (day*µg/mL)	$T_{1/2,capacity\ limited}$ (day)	$T_{1/2,non\text{-}capacity\ limited}$ (day)
800	IM	3	2.13 (0.35)	85.23 (19.43)	742.82 (291.99)	3.67 (1.09)	0.75 (0.19)
	IV	6	1.09 (2.62)	256.46 (112.25)	1833.83 (1122.46)	3.59 (1.48)	0.74 (0.32)
2000	IM	2	3.83 (2.86)	241.24 (62.58)	3733.86 (1152.57)	5.43 (1.59)	0.97 (0.56)
	IV	3	0.01 (0.00)	712.90 (149.58)	5283.24 (1177.13)	5.81 (1.39)	0.87 (0.46)

- T_{max}: IM > IV; C_{max} and AUC: IM < IV; $T_{1/2}$: IM = IV
- Non-linear pharmacokinetics (target-mediated drug disposition): higher dose exhibited decreased elimination (longer $T_{1/2}$), delayed absorption (increased T_{max}), and disproportionately higher systemic exposure

IBA PK and RO (800 mg IM)

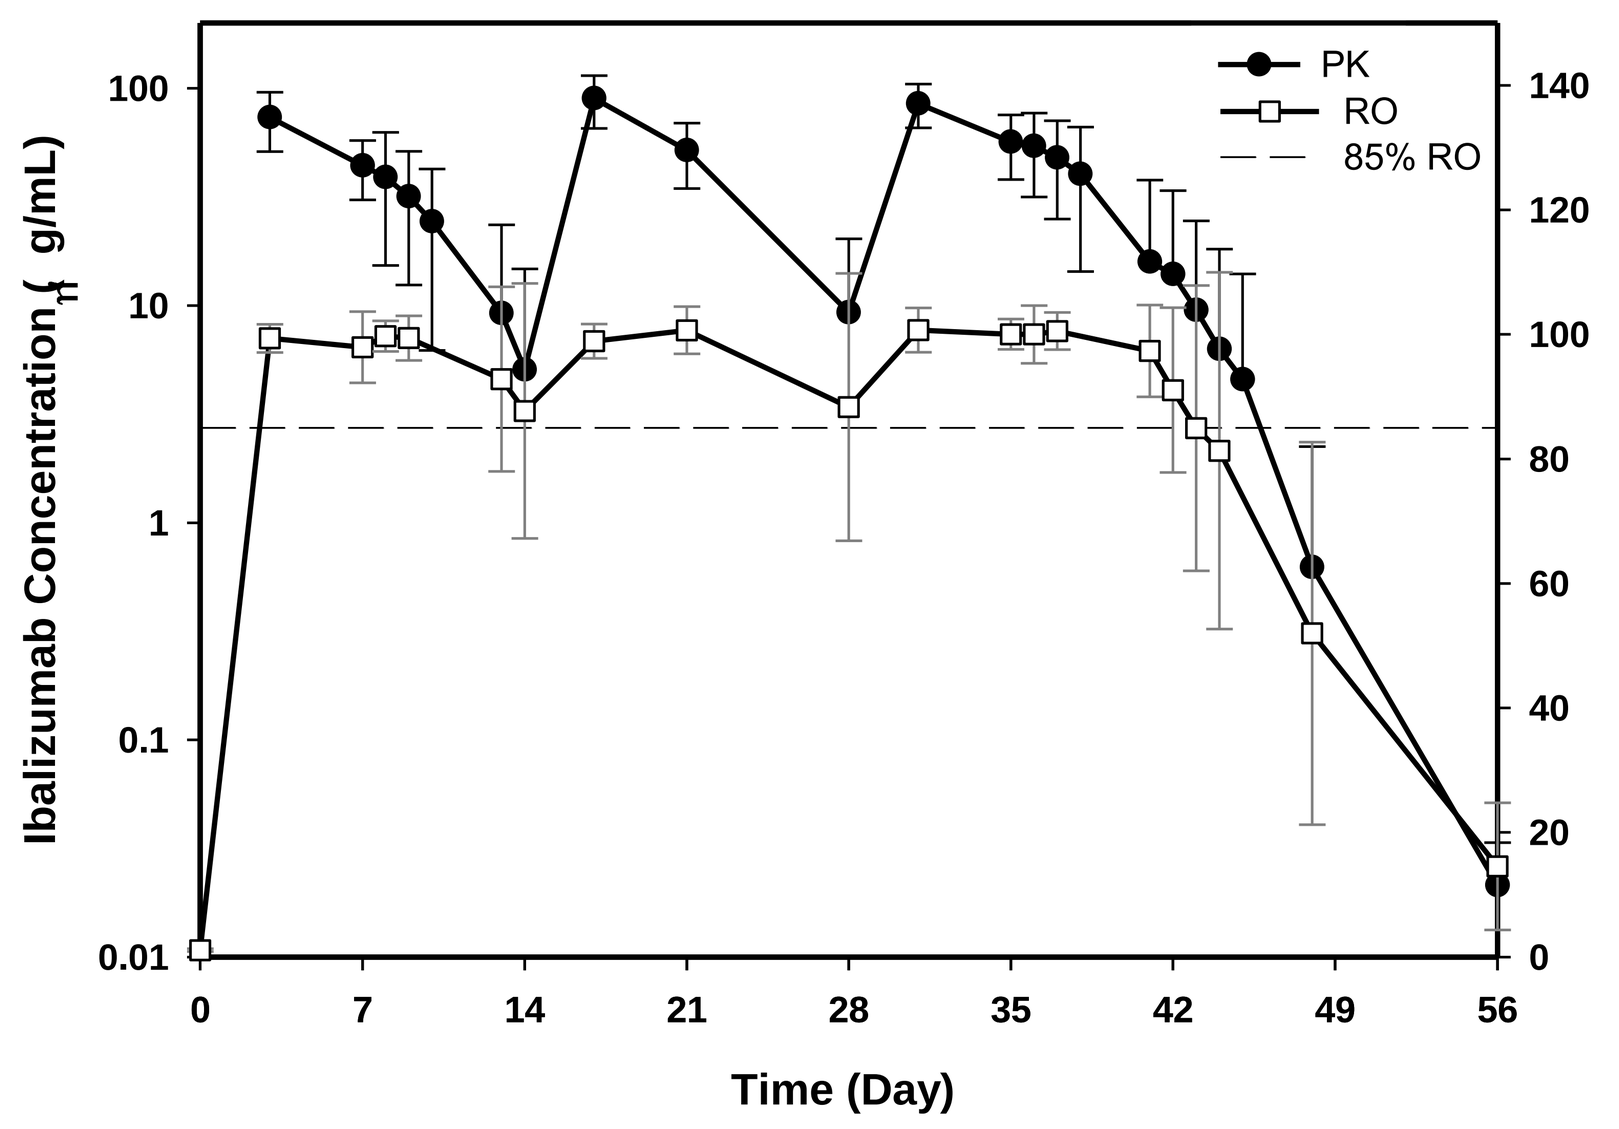
PK
RO
85% RO
Receptor Occupancy (%)
Time (Day)

IBA PK and RO (2000 mg IM)

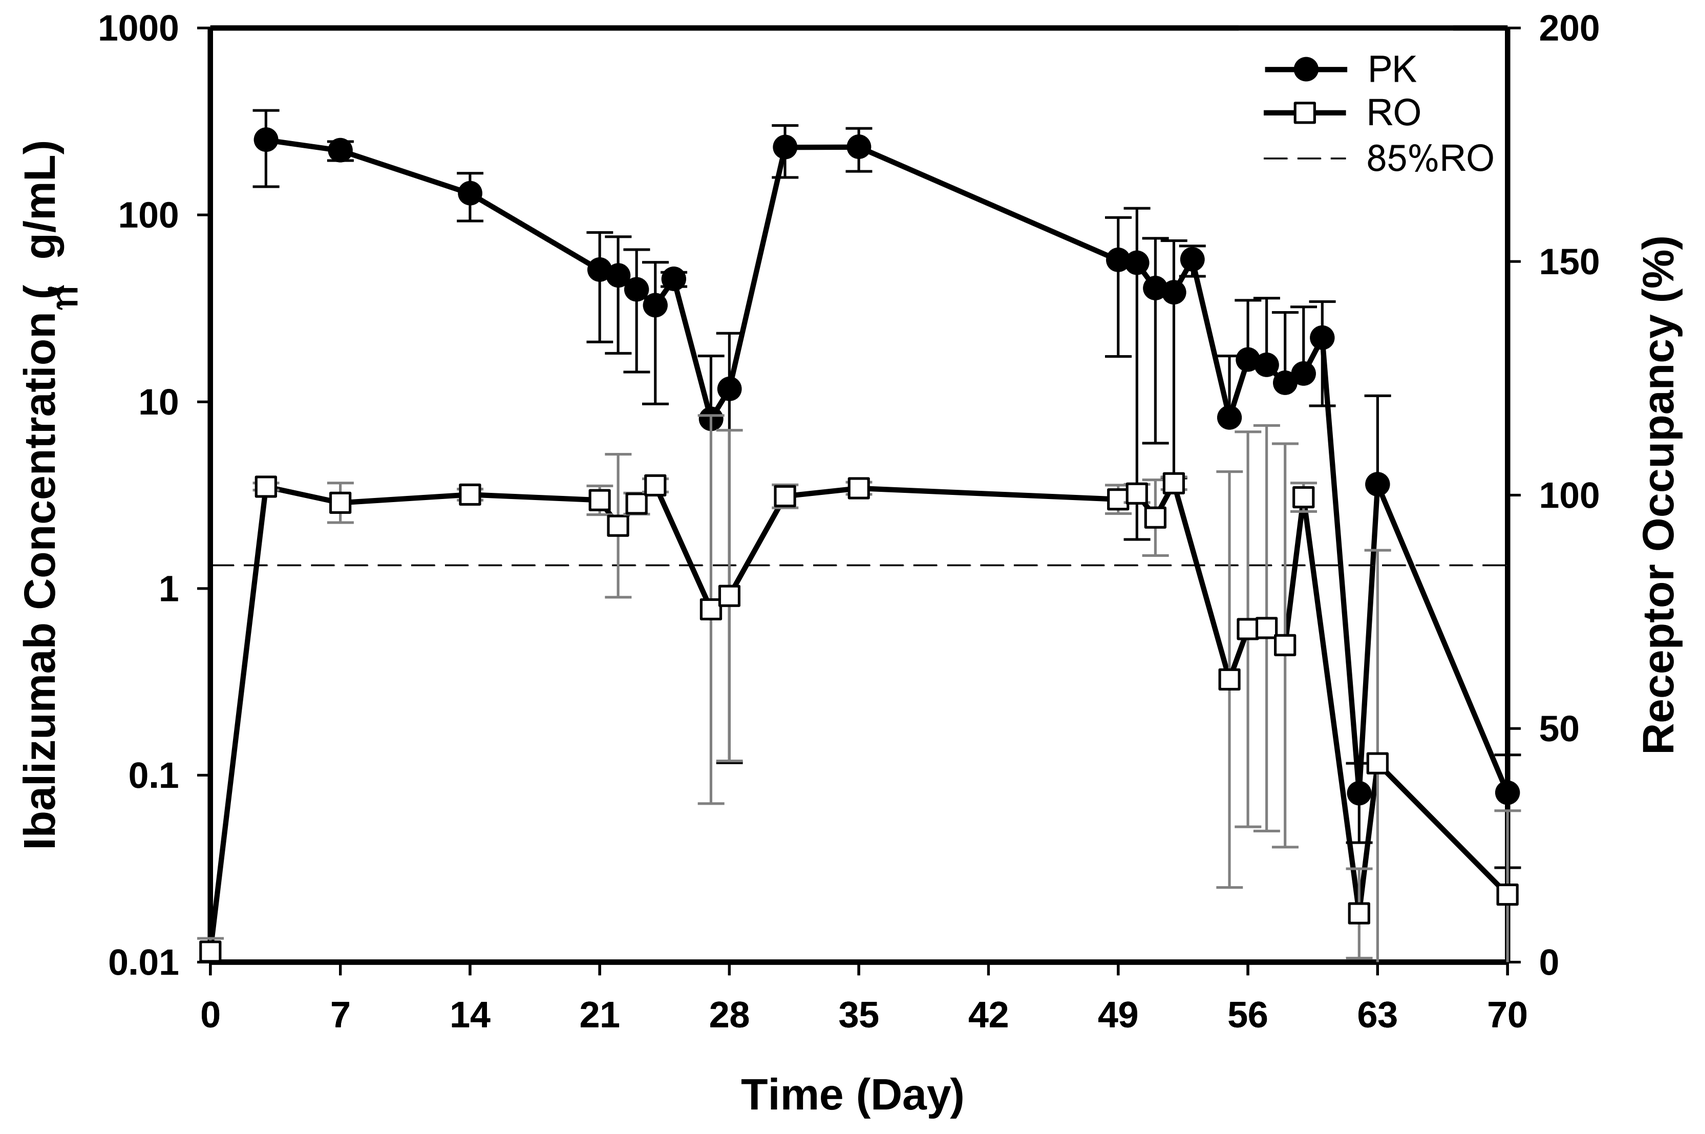
PK
RO
85%RO
Receptor Occupancy (%)
Time (Day)

- The elevation in CD4 receptor occupancy (RO) was generally associated with increased IBA concentrations.
- For the 800 mg IM dose, the mean RO was >85% during dosing period. For the 2000 mg IM dose, the mean RO was <85% but the median RO was 98% at Day 28 post-dose.

Antiviral Activity

800 mg IBA
IM (monotherapy) vs. IV (+OBR)

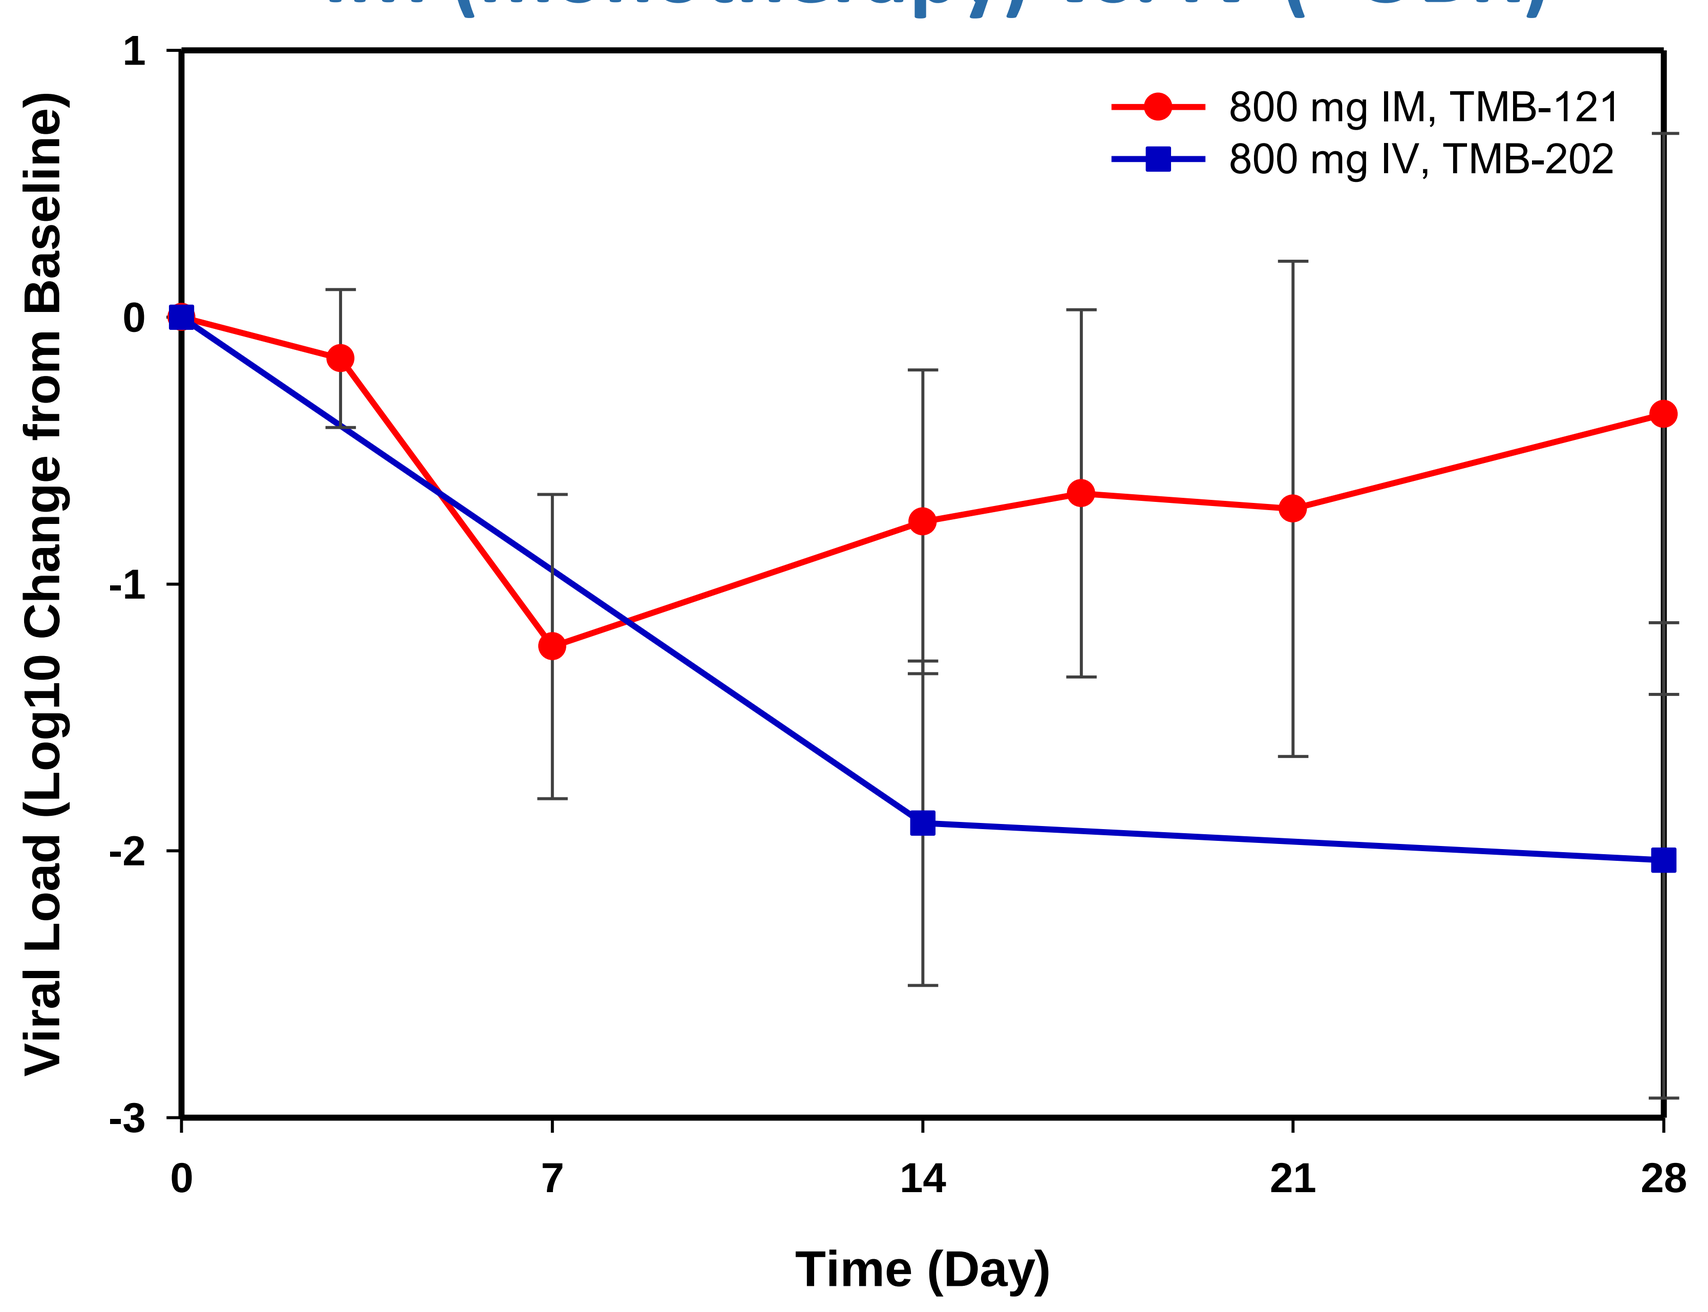
800 mg IM, TMB-121
800 mg IV, TMB-202
Viral Load (Log10 Change from Baseline)
Time (Day)

2000 mg IBA
IM (monotherapy) vs. IV (+OBR)

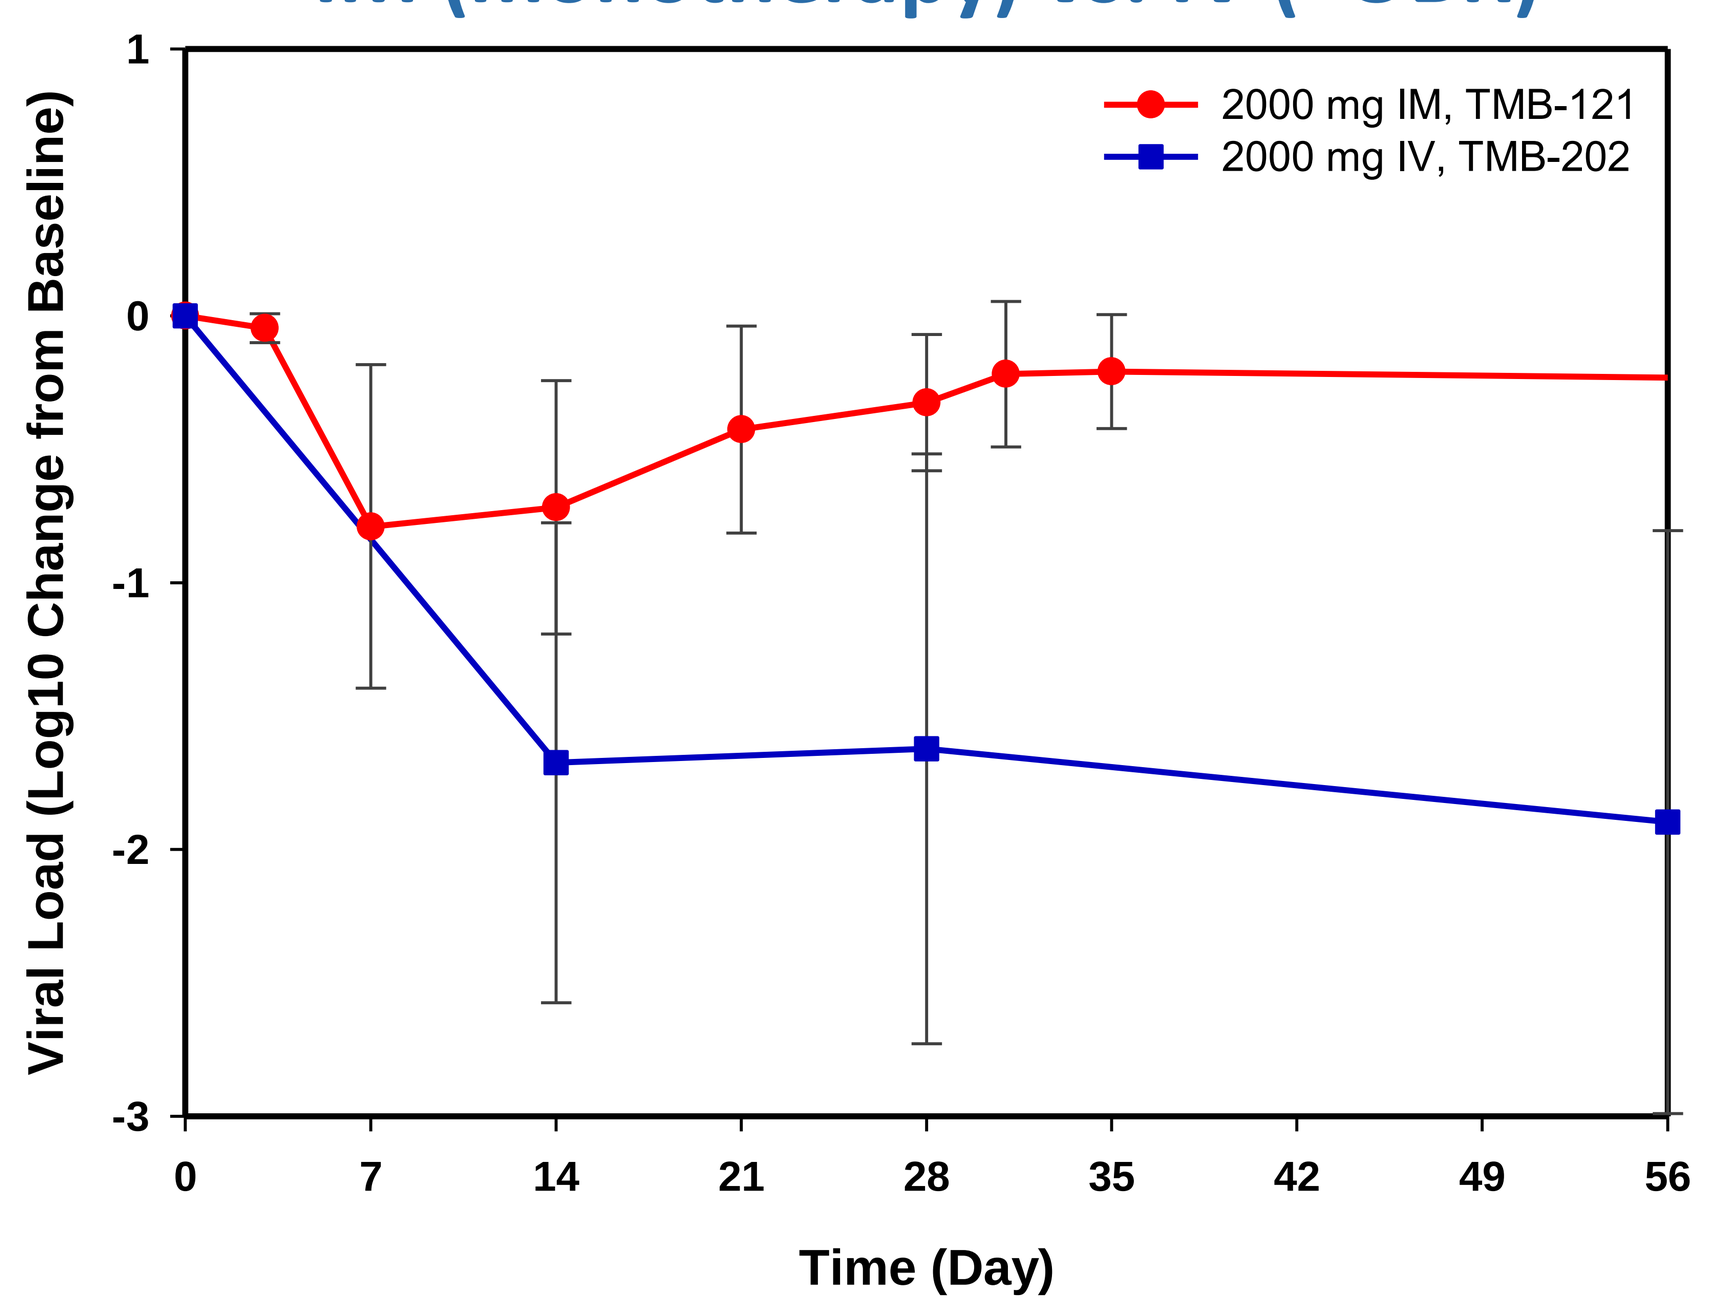
2000 mg IM, TMB-121
2000 mg IV, TMB-202
Viral Load (Log10 Change from Baseline)
Time (Day)

- **IM (monotherapy):** the maximum HIV VL reduction (1.2 $\log_{10}$ for 800 mg dose and 0.8 $\log_{10}$ for 2000 mg dose) occurred at Day 7 post-dose and rebounded to near baseline levels after 1-2 weeks. The VL rebound was likely due to resistance development with monotherapy.
- **IV (plus OBR):** the HIV VL reduction were maintained ≥ 1.0 $\log_{10}$ during the 24-week study period.

Safety and Tolerability

- **IM:** no serious adverse events (SAE) or discontinuations. No injection site reactions or anti-drug antibodies. The most frequent AE was fever.
- **IV:** 10 SAEs, none were related to study drug; 2 discontinued with AEs; 2 discontinued with AEs considered related to study drug - hypersensitivity and rash.

− The most frequent drug-related AE was rash.

Summary of Adverse Events (AEs)

Number (%) of Subjects with	TMB-121 (IM) 800 mg N=8	TMB-121 (IM) 2000 mg N=6	TMB-121 (IM) Total N=14	TMB-202 (IV) 800 mg N=59	TMB-202 (IV) 2000 mg N=54	TMB-202 (IV) Total N=113
AE(s)	5 (62.5)	2 (33.3)	7 (50.0)	50 (84.7)	44 (81.5)	94 (83.2)
AEs Related to Study Drug[1]	0 (0.0)	0 (0.0)	0 (0.0)	21 (35.6)	24 (44.4)	45 (39.8)
AE Leading to Discontinuation	0 (0.0)	0 (0.0)	0 (0.0)	2 (3.4)	1 (1.9)	3 (2.7)
Serious AE(s)	0 (0.0)	0 (0.0)	0 (0.0)	7 (11.9)	3 (5.6)	10 (8.8)
Severe AEs[2]	0 (0.0)	0 (0.0)	0 (0.0)	9 (15.3)	9 (16.7)	18 (15.9)

[1]Definitely, probably, or possibly related to study drug.
[2]Severe, life threatening, or death.

Conclusion

The PK profiles of biweekly 800 mg IBA and monthly 2000 mg IBA administered IM were comparable with IV profiles from a previous study. Both dosages of IBA administered IM were safe, well tolerated, and produced clinically significant viral load reductions at Day 7 as monotherapy. These data warrant further development of the IM administration of IBA.